Feasibility and Safety of Endoscopic Ultrasound-Guided Delivery of Human GLP-1 Pancreatic Gene Therapy in a Porcine Model

Christopher C. Thompson,[1,2] Linda S. Lee,[1,2] Jacob Wainer,[3] Michael Biasella,[3] Lindsay Schulman,[3] Nicole Picard,[3] Rebecca Reese,[3] Alice Liou Fitzpatrick,[3] Edward Mahoney,[3] Emily Cozzi,[3] Shimyn Slomovic,[3] Timothy Kieffer,[3] Jay Caplan,[3] Harith Rajagopalan[3]

[1]Brigham and Women's Hospital, Boston, Massachusetts, USA. [2]Harvard Medical School, Boston, Massachusetts, USA. [3]Fractyl Health, Inc., Burlington, Massachusetts, USA.

Introduction

- Glucagon-like peptide 1 (GLP-1) therapy is now the cornerstone of metabolic disease treatment. Although efficacious, discontinuation is frequent and results in near-complete glycemic and weight rebound
- We have developed a single-dose, pancreatic gene therapy (PGTx) that enables durable, nutrient-responsive, islet-driven GLP-1 production (Smart GLP-1™) (**Figure 1.**)
- In murine models, we have previously shown that GLP-1 PGTx can improve glucose control, induce weight loss, and durably maintain weight loss after semaglutide withdrawal

Here, we evaluate the feasibility and safety of delivering human GLP-1 PGTx (RJVA-001) with an endoscopic ultrasound (EUS)-guided delivery system into porcine pancreas (Figure 2.)

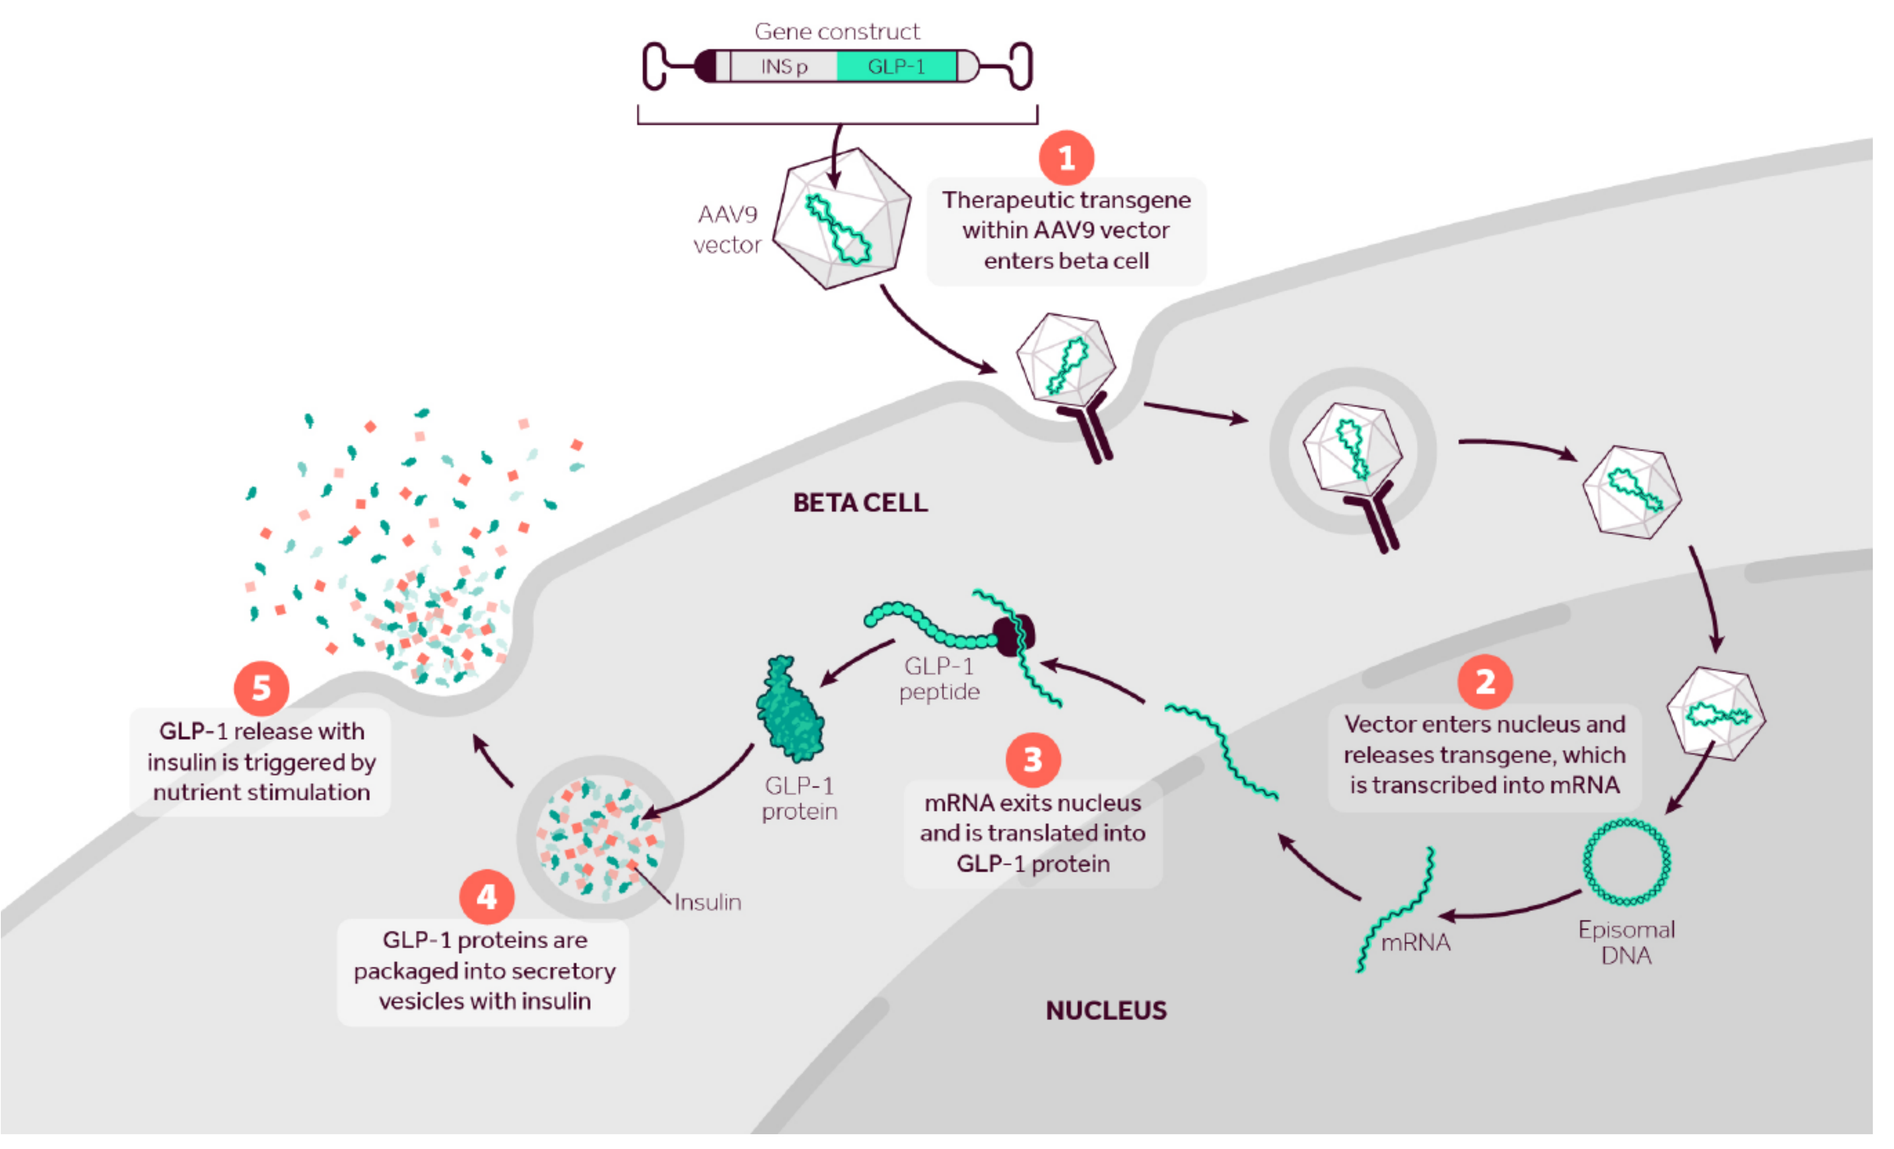
Gene construct
INS p
GLP-1
AAV9 vector
1 Therapeutic transgene within AAV9 vector enters beta cell
BETA CELL
2 Vector enters nucleus and releases transgene, which is transcribed into mRNA
Episomal DNA
mRNA
3 mRNA exits nucleus and is translated into GLP-1 protein
GLP-1 peptide
GLP-1 protein
Insulin
4 GLP-1 proteins are packaged into secretory vesicles with insulin
5 GLP-1 release with insulin is triggered by nutrient stimulation
NUCLEUS

Figure 1. Human GLP-1 PGTx (RJVA-001) Therapeutic Mechanism of Action. **1**) The RJVA-001 transgene construct, consisting of human insulin promoter and GLP-1 sequences, is packaged into adeno-associated virus (AAV)-9 vectors which are taken up by the beta cell. **2**) Vectors enter the nucleus and release the transgene, which is transcribed into GLP-1 mRNA. **3**) GLP-1 mRNA exits the nucleus and is translated into protein. **4**) GLP-1 proteins are packaged into secretory vesicles with insulin. **5**) GLP-1 with insulin release is triggered by nutrient stimulation.

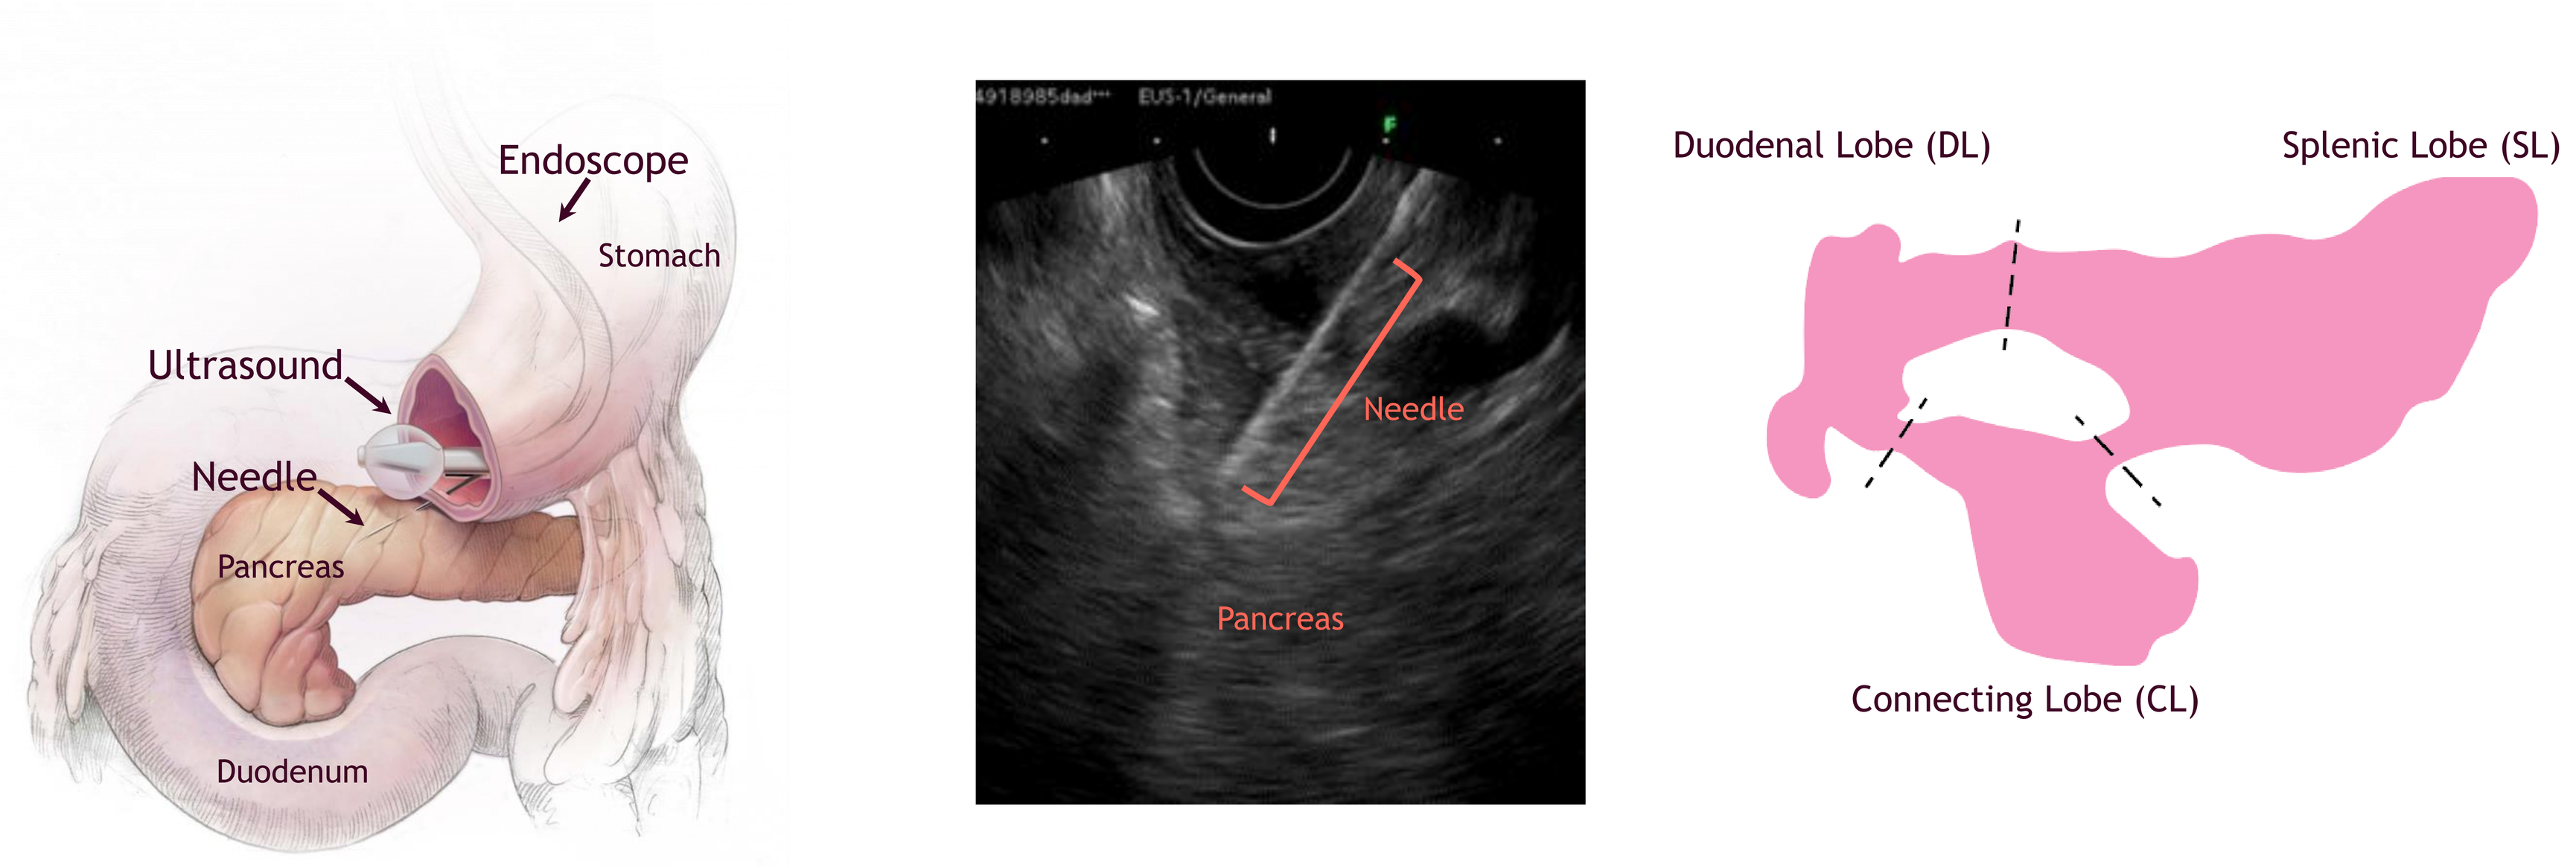
A)
Endoscope
Stomach
Ultrasound
Needle
Pancreas
Duodenum
B)
EUS-1/General
Needle
Pancreas
C)
Duodenal Lobe (DL)
Splenic Lobe (SL)
Connecting Lobe (CL)

Figure 2. EUS-Guided Delivery System and Pancreatic Infusion Procedure. An echoendoscope with a custom needle integrated with a proprietary fluid delivery system is tracked into the porcine stomach. Utilizing ultrasound, anatomic landmarks (splenic artery, spleen, and liver) guide transgastric infusions of RJVA-001 into the parenchymal tissue (**A** and **B**) of the targeted splenic lobe of the porcine pancreas (**C**, equivalent to the human body and tail of the pancreas).

Methods and Results

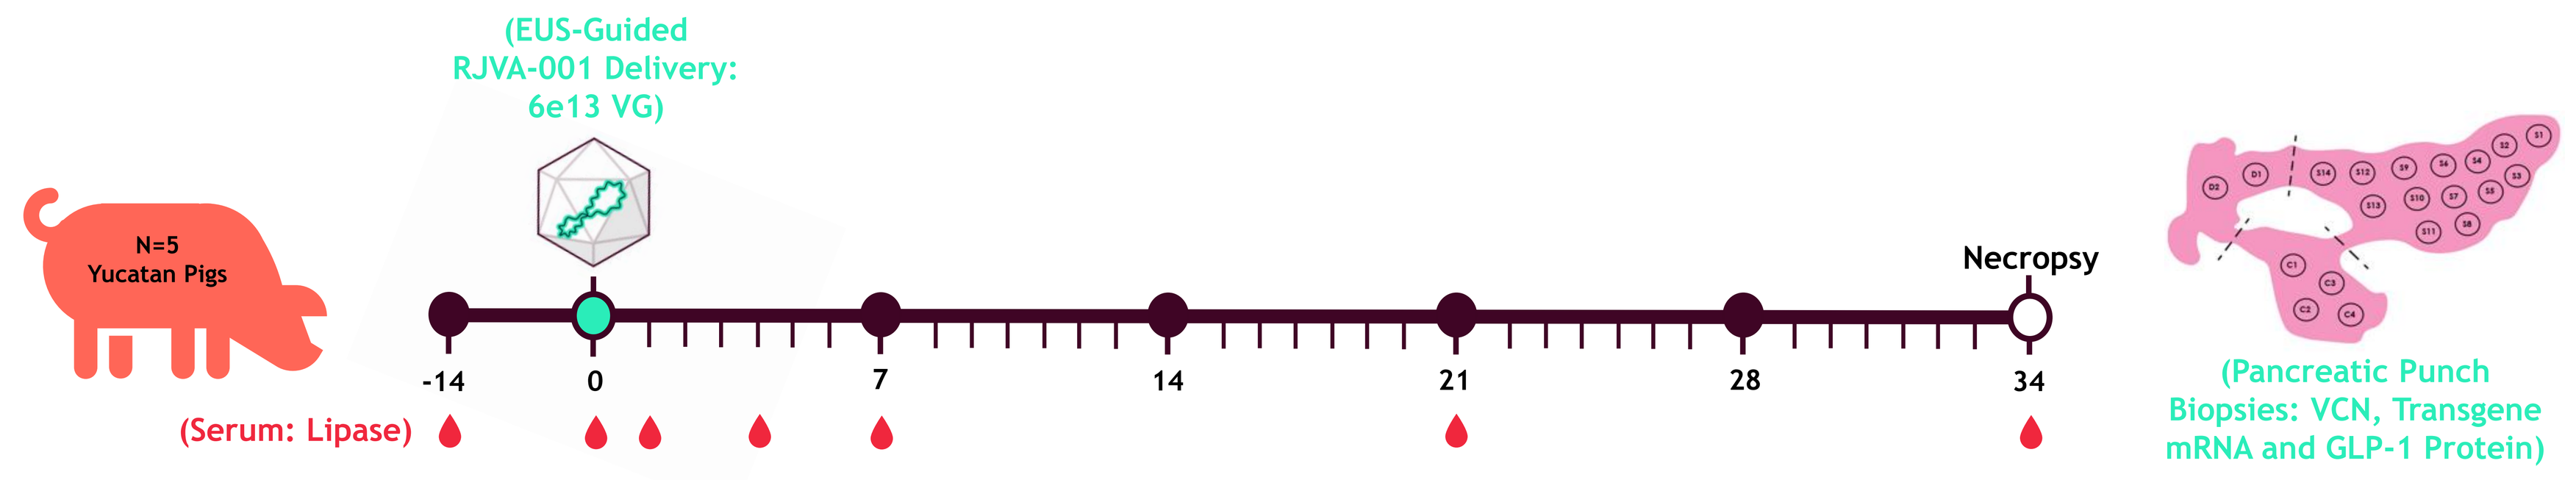
N=5 Yucatan Pigs
(EUS-Guided RJVA-001 Delivery: 6e13 VG)
-14
0
7
14
21
28
34
Necropsy
(Serum: Lipase)
(Pancreatic Punch Biopsies: VCN, Transgene mRNA and GLP-1 Protein)

Figure 3. Porcine Study Design and Analyses. RJVA-001 [6e13 vector genome (VG)] EUS-guided delivery was evaluated in five non-diseased Yucatan pigs. Serum lipase, as a surrogate marker of pancreatitis, was assessed on days -14, 0, 1, 4, 7, 21, and 34 to establish early and longer-term procedural safety. Animals were necropsied at day 34 post-procedure and punch biopsies were taken from 20 locations across the pancreas to establish vector copy number (VCN), transgene ribonucleic acid (RNA) and protein expression as indicators of vector transduction efficiency and RJVA-001-driven GLP-1 production.

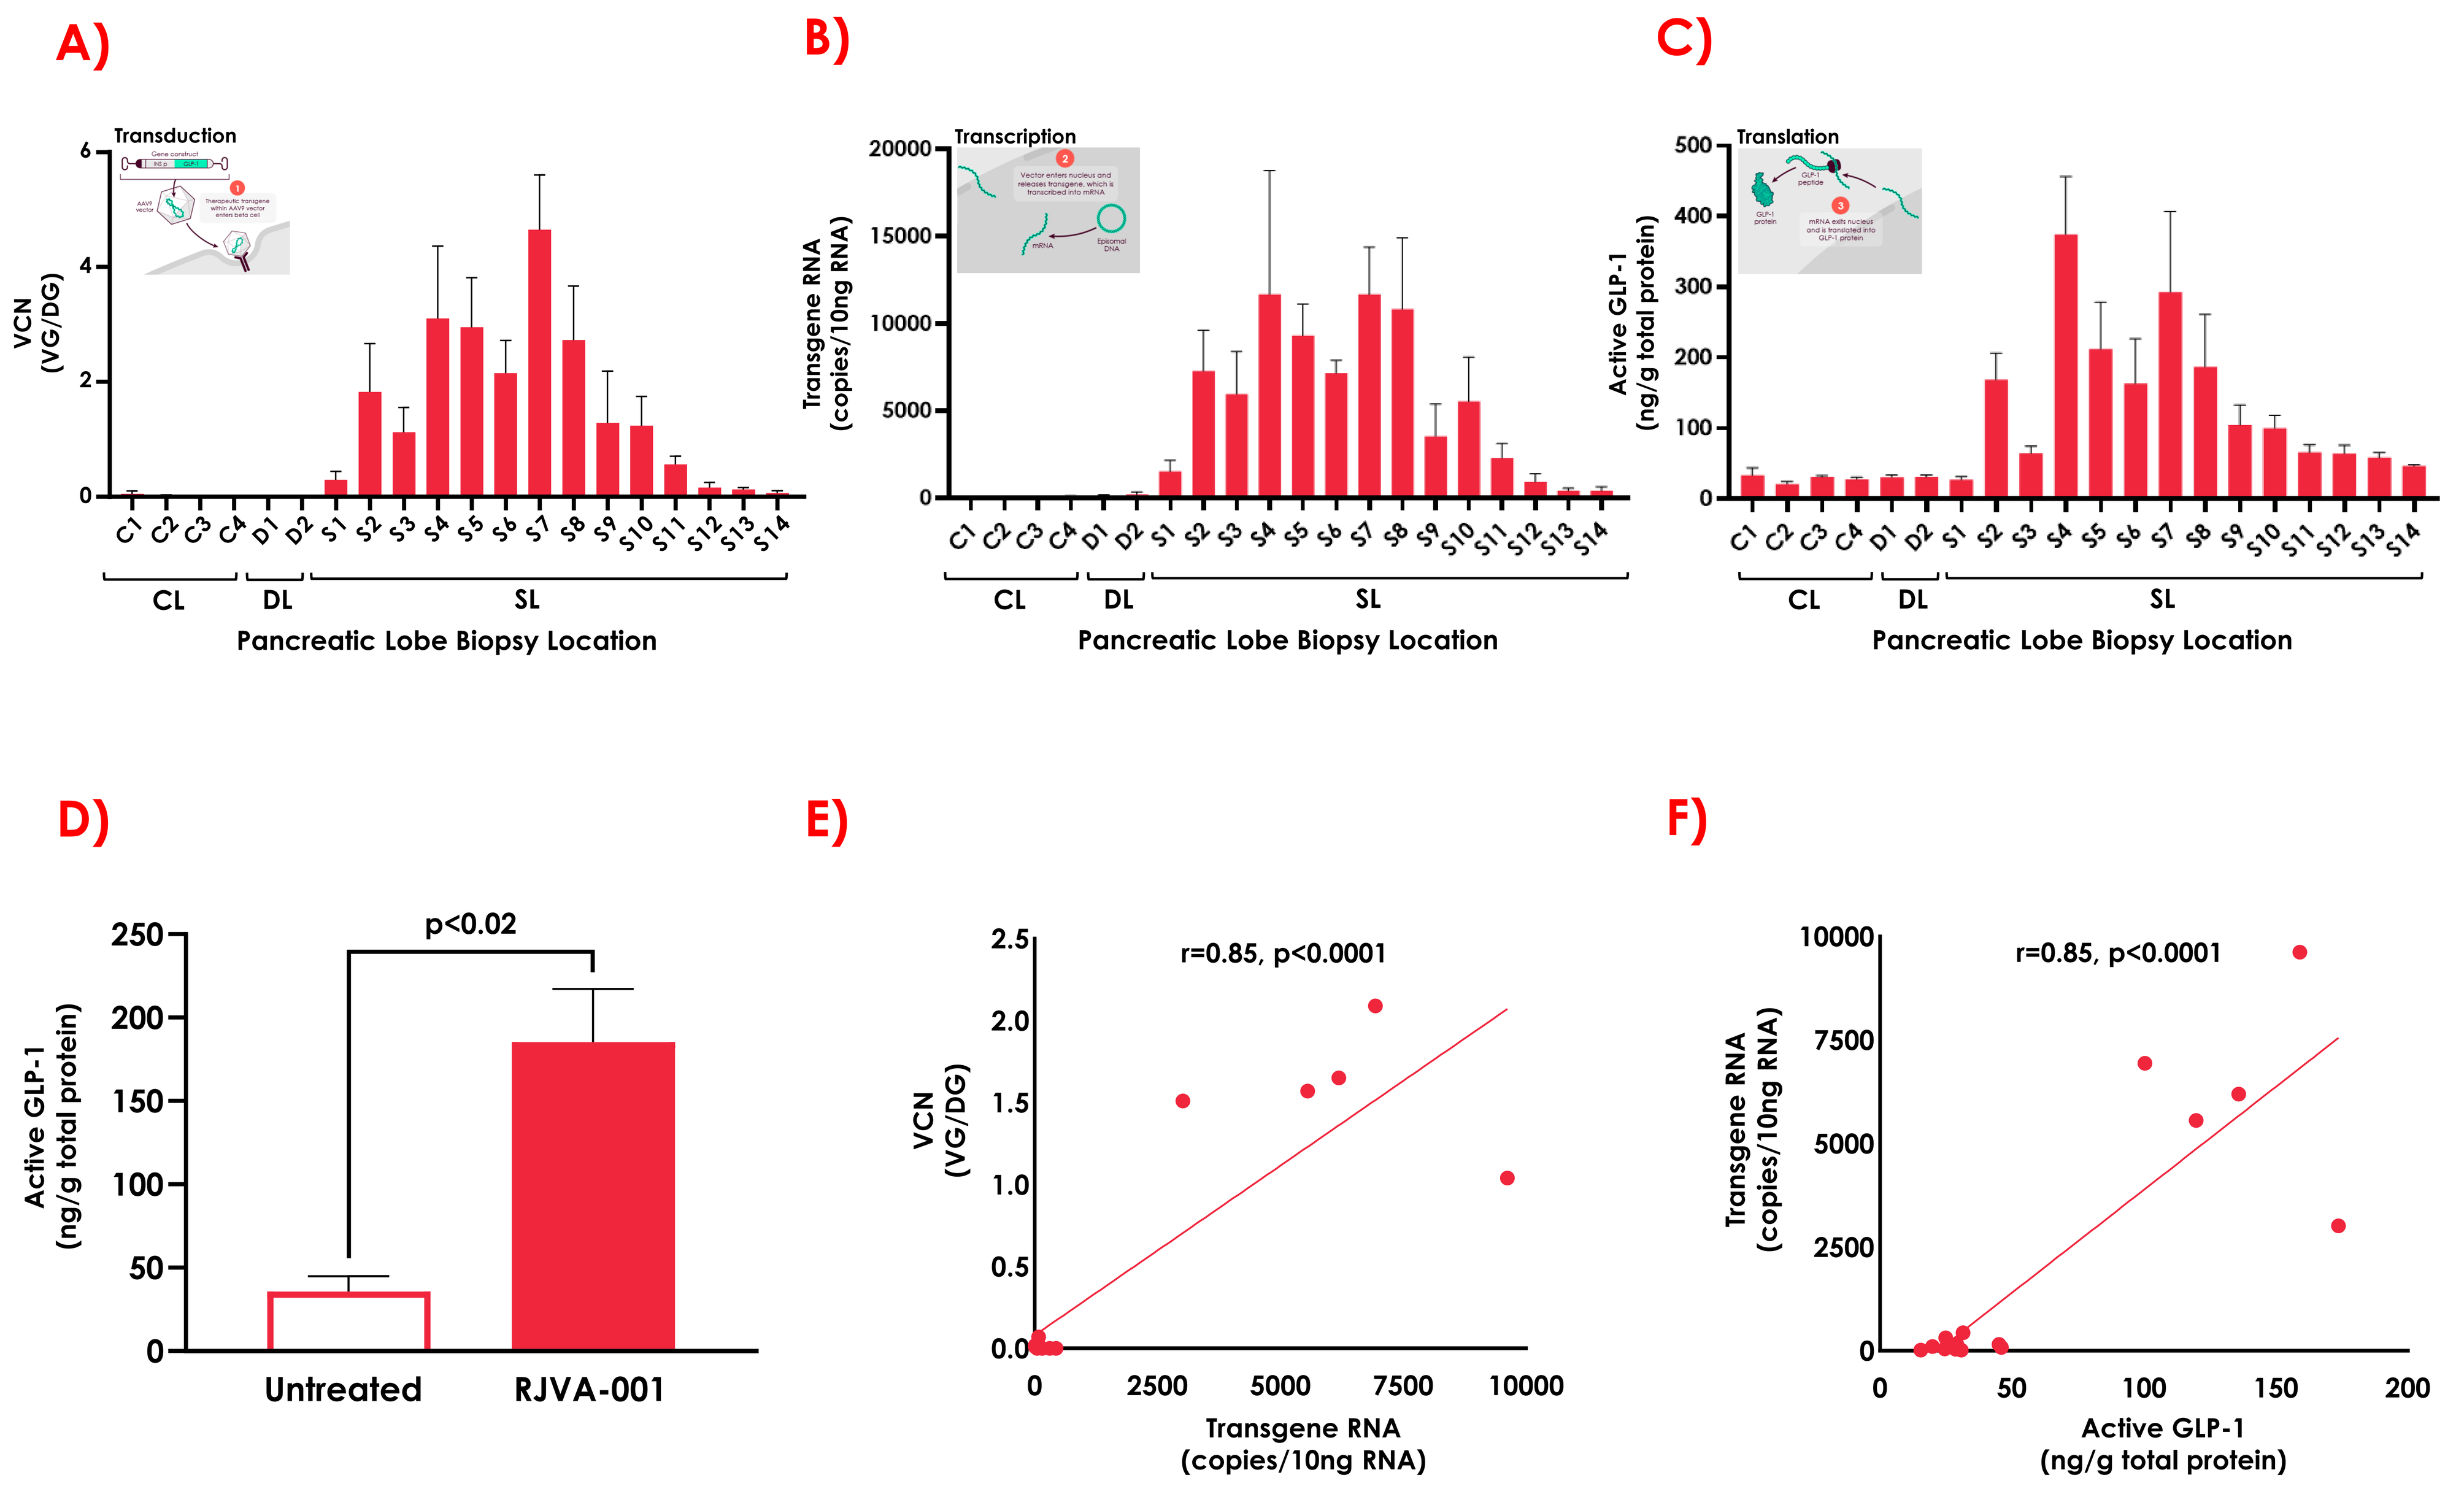
A)
Transduction
VCN (VG/DG)
B)
Transcription
Transgene RNA (copies/10ng RNA)
C)
Translation
Active GLP-1 (ng/g total protein)
C1 C2 C3 C4 D1 D2 S1 S2 S3 S4 S5 S6 S7 S8 S9 S10 S11 S12 S13 S14
CL
DL
SL
Pancreatic Lobe Biopsy Location
D)
p<0.02
Active GLP-1 (ng/g total protein)
Untreated
RJVA-001
E)
r=0.85, p<0.0001
VCN (VG/DG)
Transgene RNA (copies/10ng RNA)
F)
r=0.85, p<0.0001
Transgene RNA (copies/10ng RNA)
Active GLP-1 (ng/g total protein)

Figure 4. EUS-Guided RJVA-001 Delivery Led to Robust AAV9 Transduction, GLP-1 Transcription, and a Five-Fold Increase in Active Pancreatic GLP-1 Protein. Transgastric delivery of RJVA-001 resulted in increased adeno-associated virus (AAV)-9 transduction [**A**, shown as vector genome per diploid genome(VG/DG)], GLP-1 transgene transcription (**B**), and subsequent active GLP-1 protein expression (**C**) within the targeted splenic lobe of the pancreas. Active GLP-1 protein was five-fold greater in RJVA-001 treated animals compared to untreated controls (**D**, $p<0.02$). Pancreatic vector copy number (VCN), and RJVA-001 transgene RNA levels were positively correlated (**E**, $r=0.85$, $p<0.0001$). Likewise, RNA levels correlated with RJVA-001 islet-driven GLP-1 protein levels (**F**, $r=0.85$, $p<0.0001$). Data shown as mean ± standard error of the mean (A-D), mean lobe values (E and F). CL=connecting lobe, DL=duodenal lobe, SL=splenic lobe.

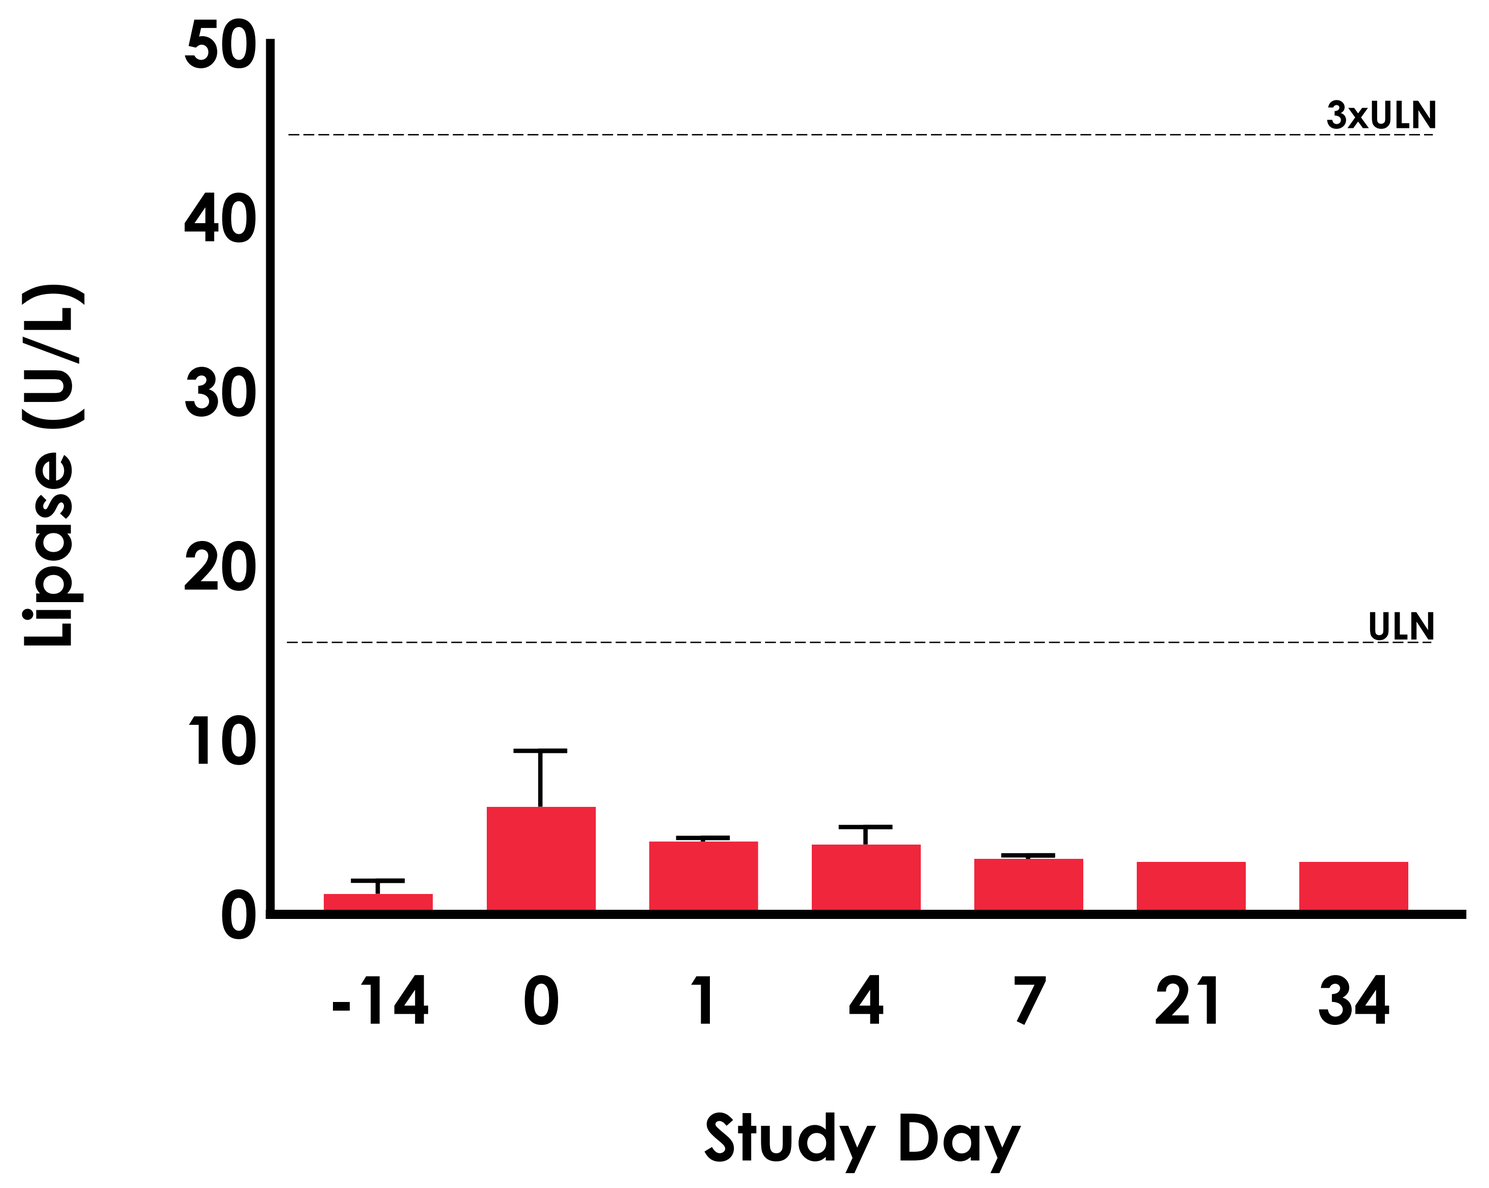
Lipase (U/L)
3xULN
ULN
-14
0
1
4
7
21
34
Study Day

Figure 5. EUS-Guided RJVA-001 Delivery Was Well-Tolerated. All 5 animals tolerated the transgastric delivery procedure well with serum lipase levels remaining within the upper limit of normal (ULN) at all timepoints evaluated from day -14 pre-procedure to day 34 post-procedure. Data shown as mean ± standard error of the mean.

Conclusions and Next Steps

These data demonstrate that RJVA-001 can be safely delivered endoscopically in a large animal model allowing for direct pancreatic targeting

RJVA-001-driven human pancreatic GLP-1 production demonstrates the feasibility of single-dose local gene therapy as a durable strategy for the treatment of metabolic diseases

Anticipate submitting the first clinical trial application module for RJVA-001 in type 2 diabetes in the first half of 2025

Publications and Presentations

Fractyl.com

Digestive Disease Week®

Disclaimer: Pancreatic gene therapy (PGTx) is a preclinical development program that has yet to be assessed by regulatory bodies for investigational or commercial use.
Disclosures: CCT is a consultant and serves on advisory boards for Fractyl Health, Inc. He is also a shareholder of Fractyl Health, Inc. LSL is a consultant for Fractyl Health, Inc TK, JW, MB, LS, NP, RR, AF, EC, SS, JC & HR are employees and share or option holders of Fractyl Health, Inc. HR is also a board member of Fractyl Health, Inc. EM is a former employee of Fractyl Health, Inc.
Acknowledgments: The authors thank CBSET for porcine preclinical research services.